Phase I study to assess biodistribution of CB307, a trispecific Humabody® targeting CD137, prostate- specific membrane antigen, and human serum albumin with 89Zr-CB307 PET

ESMO 2024 - Barcelona

Poster number

667P

Knapen DG[1], van Winkel CAJ[2], Grevelink T[1], van Sluis J[3], Lub-de Hooge MN[2], Brouwers AH[3], van der Vegt M[3], Pizzey J[4], Williams R[4], Archer S[4], Bland-Ward PA[4], Duffy K[4], Hashimoto K[4], de Groot DJA[1], de Vries EGE[1]

1 Department of Medical Oncology, University Medical Center Groningen, University of Groningen, Groningen, the Netherlands
2 Clinical Pharmacy and Pharmacology, University Medical Center Groningen, University of Groningen, Groningen, the Netherlands
3 Medical Imaging Center, University Medical Center Groningen, University of Groningen, Groningen, the Netherlands
4 Crescendo Biologics Ltd, Babraham Research Campus, Cambridge, United Kingdom

Background

CB307, a trispecific Humabody® targeting CD137 (4-1BB), prostate-specific membrane antigen (PSMA), and human serum albumin, lacks an Fc domain and weighs < 50 kDa. It was designed to activate T cells exclusively in PSMA+ lesions. Preclinical studies have confirmed efficient CB307 delivery to PSMA+ tumors in mice. To understand its biodistribution, patients were imaged with PET after injection with 89-zirconium (^{89}Zr) labeled CB307.

Aim: To define the behavior of CB307 in patients with PSMA-positive tumors.

Fig. 1: Schematic overview of CB307 molecular structure and suggested mode of action.
A) CB307 consists of three peptide-linked Humabody® V_H domains. B) The CB307 design allows CD137 clustering and subsequent T cell co-stimulation in the presence of PSMA high-expressing tumor cells. C) In PSMA low-expressing (normal) cells, CD137 clustering is prevented, therefore not resulting in T cell co-stimulation.

PSMA, prostate-specific membrane antigen

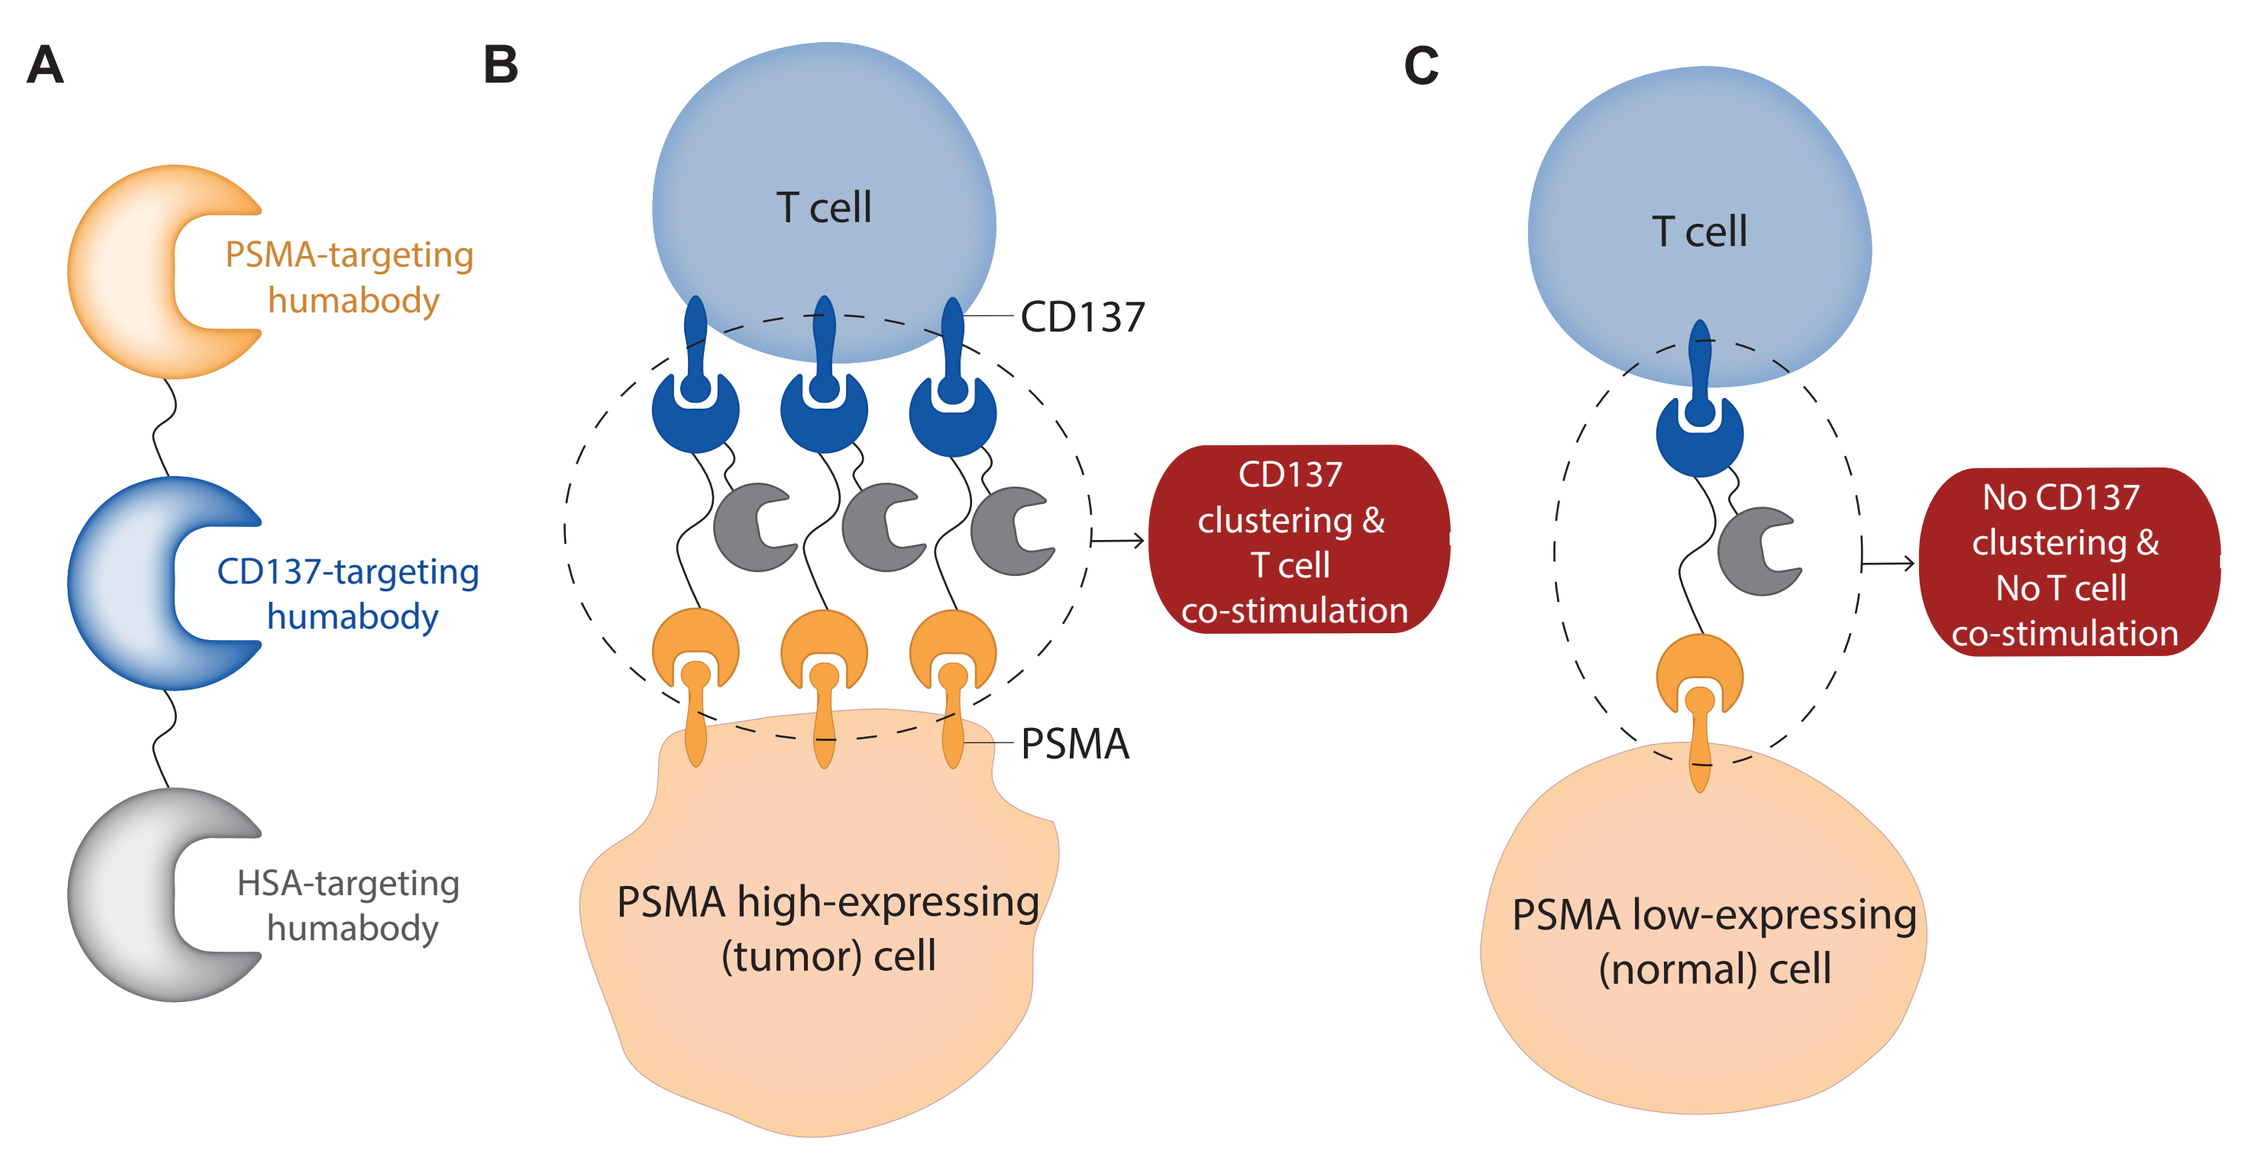
A
B
C
PSMA-targeting humabody
CD137-targeting humabody
HSA-targeting humabody
T cell
CD137
PSMA
CD137 clustering & T cell co-stimulation
PSMA high-expressing (tumor) cell
T cell
No CD137 clustering & No T cell co-stimulation
PSMA low-expressing (normal) cell

Methods

Patients with PSMA-positive tumors underwent a PSMA-PET scan at screening and received ^{89}Zr-CB307 (37 MBq / 5 mg protein) at Day 0 and escalating doses of unlabeled CB307 up to a maximum of 100 mg to achieve sufficient tracer in the circulation to allow tumor uptake. Subsequently, patients underwent 3 PET scans (Day 2, 5 and 7) followed by a tumor biopsy (NCT05836623) on Day 7. Thereafter, patients entered the CB307 treatment trial (NCT04839991). Tracer uptake was measured as standardized uptake value in tumor lesions (SUVmax) and healthy tissues (SUVmean). Blood tracer pharmacokinetics and urine tracer concentration were assessed. Tumor biopsies were stained with hematoxylin-eosin and immunohistochemically for PSMA, and autoradiography was performed. With anti-albumin magnetic beads, the % tracer bound to albumin was assessed. Tracer integrity was determined with SDS-PAGE.

Trial design

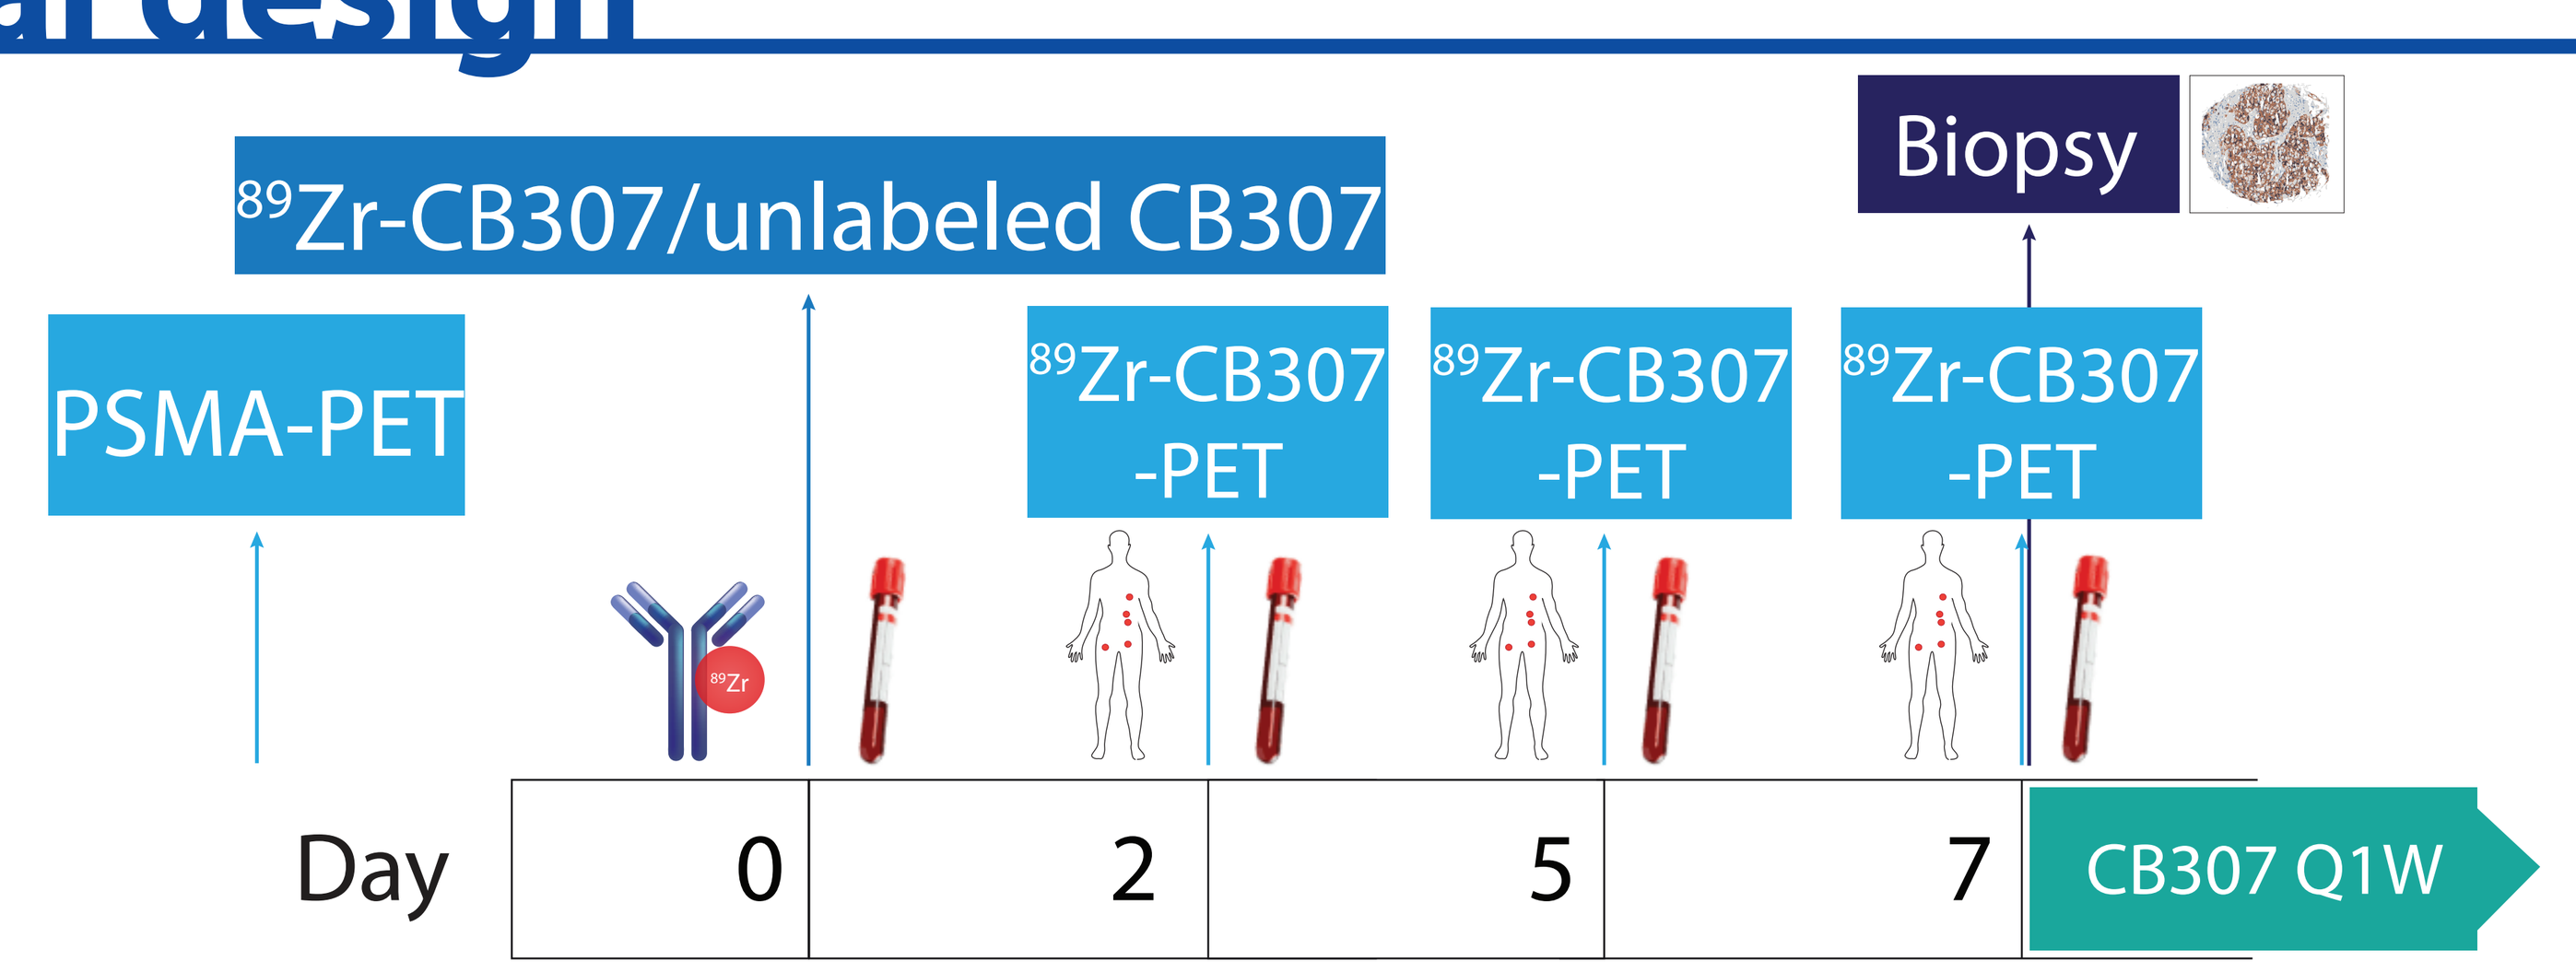
Biopsy
89Zr-CB307/unlabeled CB307
PSMA-PET
89Zr-CB307 -PET
89Zr-CB307 -PET
89Zr-CB307 -PET
Day
0
2
5
7
CB307 Q1W

Fig. 2: Trial Design
PSMA, prostate-specific membrane antigen; PET, positron emission tomography; 89Zr, 89-Zirconium; Q1W, once per week

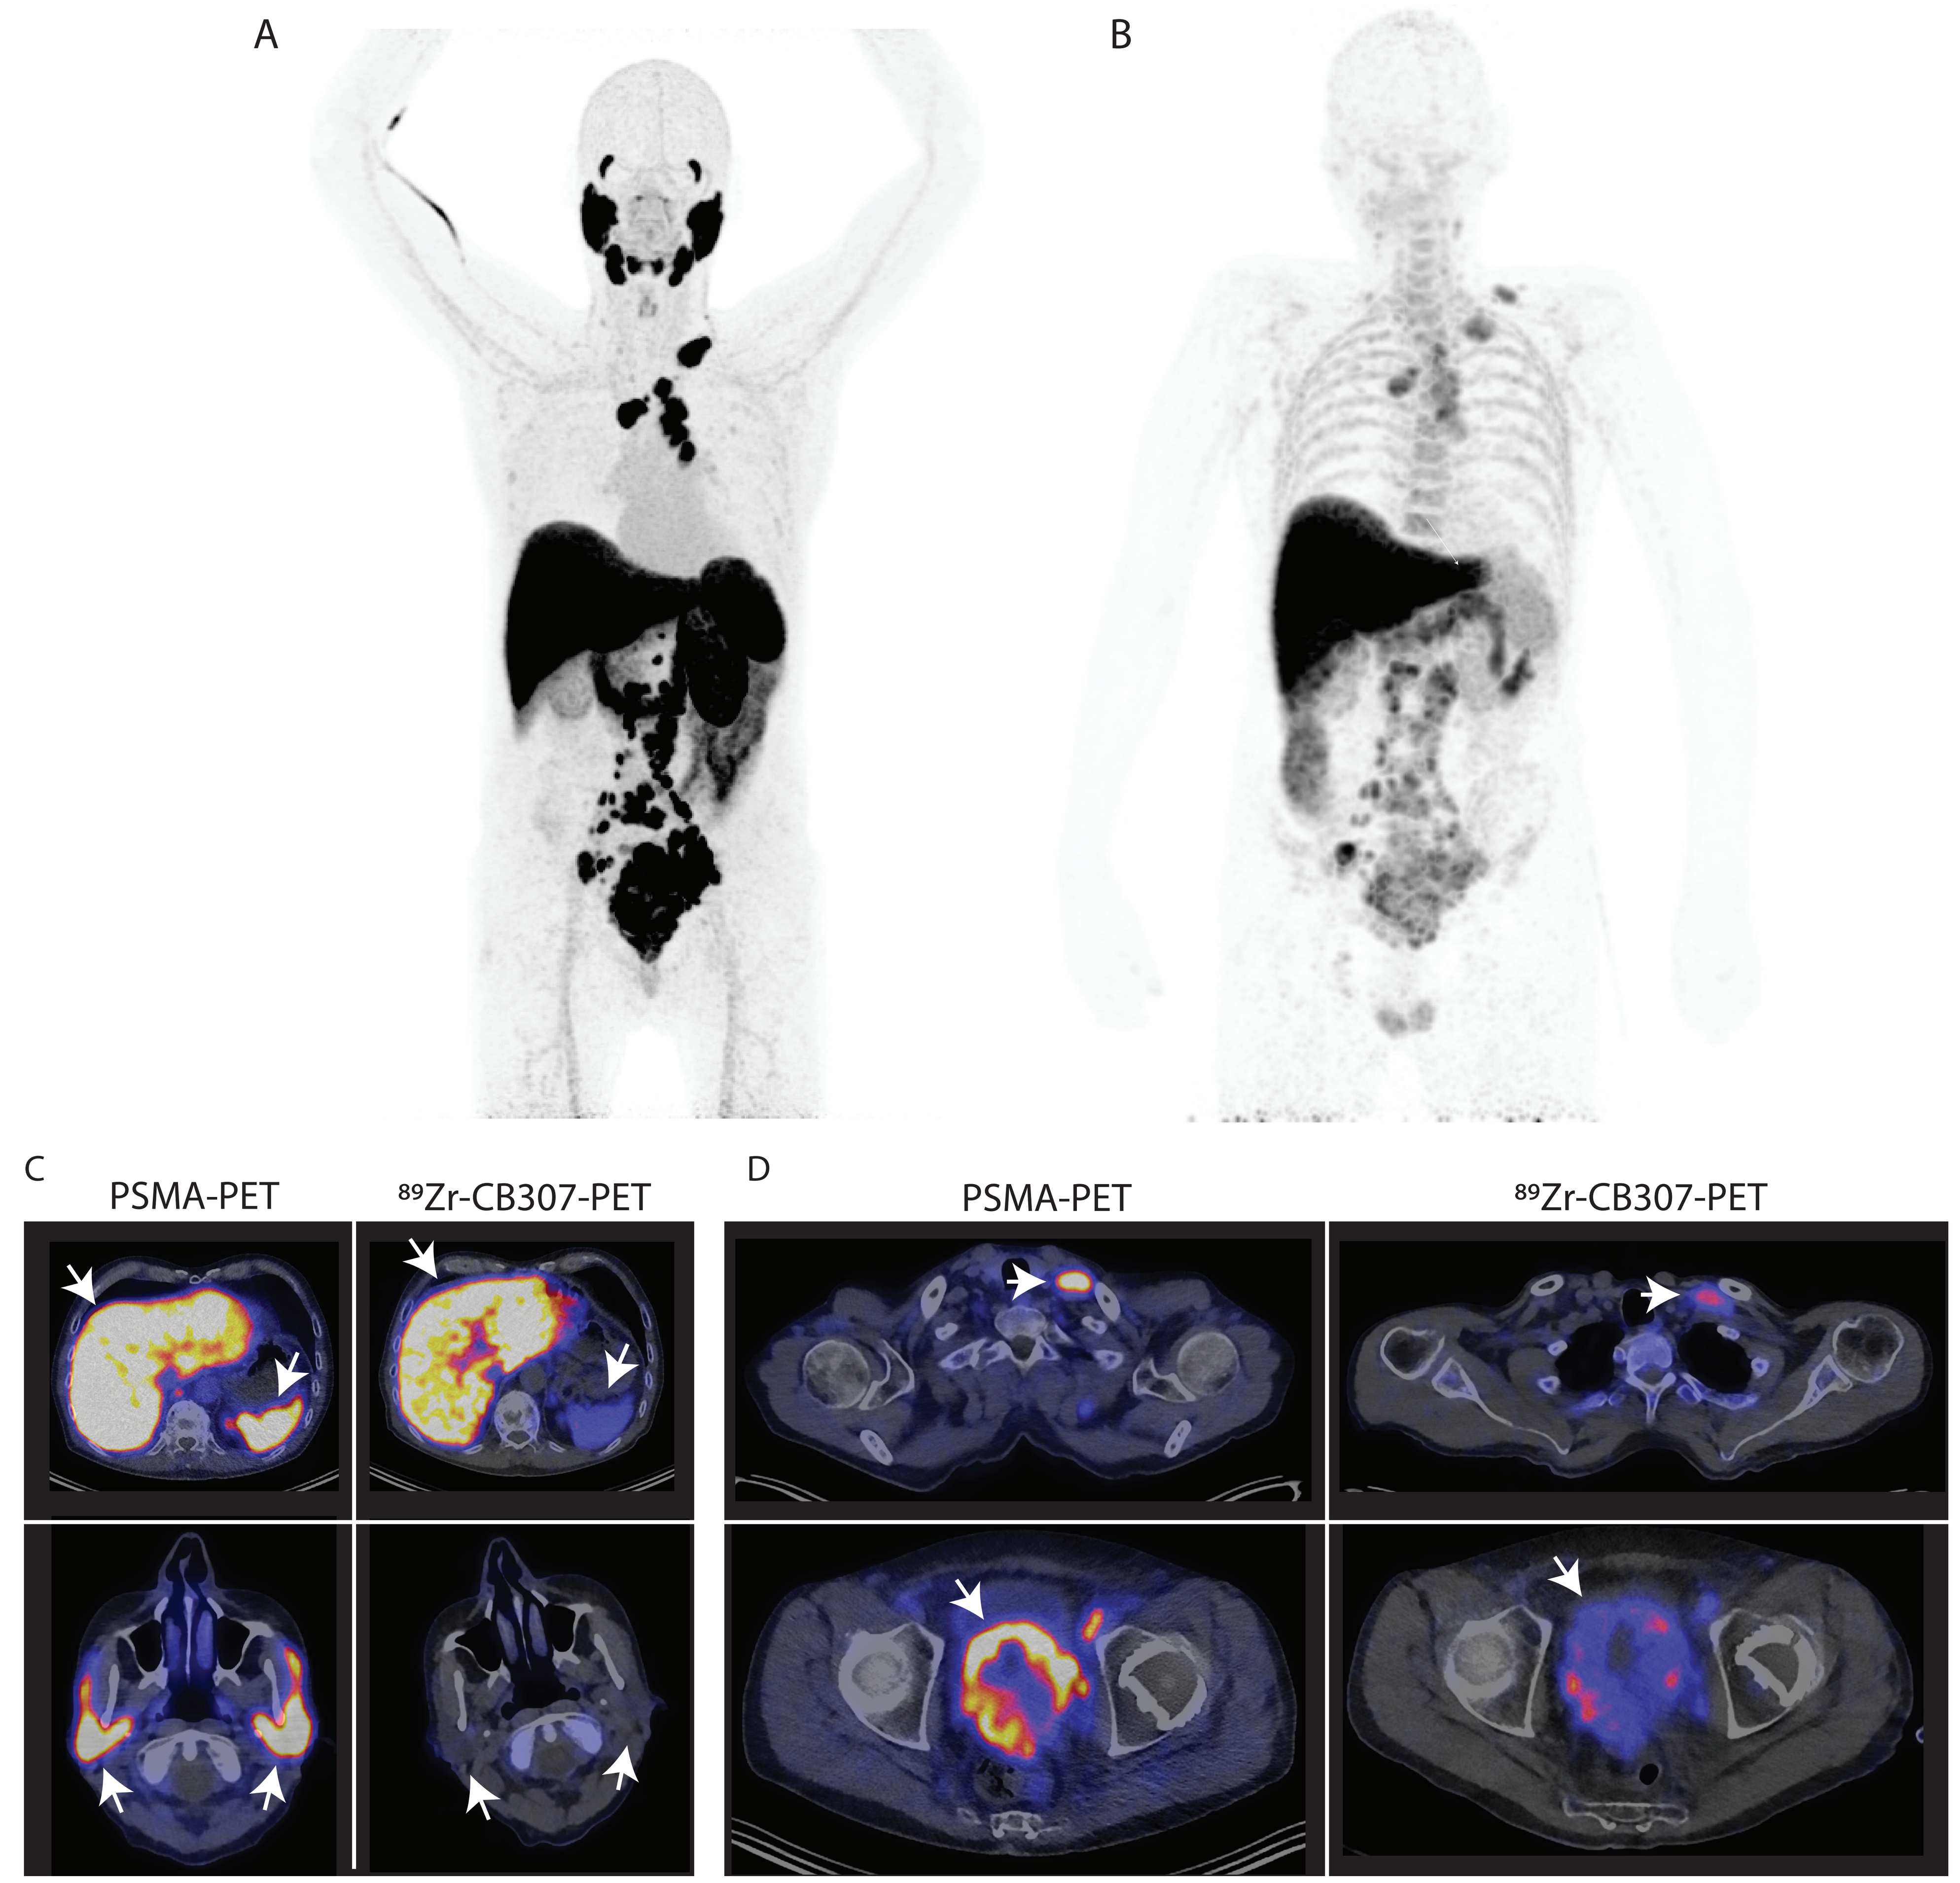
A
B
C
PSMA-PET
89Zr-CB307-PET
D
PSMA-PET
89Zr-CB307-PET

Fig. 3: Normal tissue and tumor biodistribution of ^{89}Zr-CB307
Representative MIPs of a patient with mCRPC with A) PSMA-PET and B) ^{89}Zr-CB307-PET. C) Representative PET images showcasing no specific uptake in spleen and salivary glands in the ^{89}Zr-CB307-PET. D) Representative PET images demonstrating specific uptake in PSMA-postive tumor lesions in the ^{89}Zr-CB307-PET.

PET, positron emission tomography; PSMA, prostate-specific membrane antigen; mCRPC, castration-resistant PSMA positive prostate cancer; MIP, maximum intensity projection; ^{89}Zr, 89-Zirconium;

Results

Five patients were enrolled. No tracer-related adverse events were reported. One patient received 5 mg ^{89}Zr-CB307 without unlabeled CB307, one 25 mg unlabeled CB307, and three 100 mg unlabeled CB307 with PET/CT scans at 2, 5, and 7 days (±1). The blood ^{89}Zr-CB307 T½ was 67.1 h at 105 mg. At 105 mg, optimal tumor-to-blood ratio (10.3) occurred on day 7, and the mean tumor lesion (n=18) SUVmax was 5.8 ± SD 2.1. ^{89}Zr-CB307 SUVmean in liver was 13.7 ± SD 1.3 and spleen 3.1 ± SD 0.4, without specific uptake in lymph nodes, bone marrow, and salivary glands. Tumor biopsies showed radioactive signal. Circulating tracer was intact and ~100% albumin-bound, urine ^{89}Zr-radioactivity was <2.0% of the injected dose in the 8 h post-injection, and SDS page results showed only unconjugated ^{89}Zr in urine.

Fig. 4: Tumor tissue IHC analysis and colocalization with ^{89}Zr-CB307 radioactivity.

Biopsies collected after the last PET scan were analyzed with autoradiography. Whole formalin-fixed paraffin-embedded (FFPE) blocks were exposed for 7 days with 2 µL of 1%, 0.1%, 0.05%, 0.01%, and 0.005% injected tracer on silica gel on thin-layer chromatography Al foils (Sigma-Aldrich) to an imaging plate (BAS-MS; Fujifilm) and captured with a Typhoon laser scanner (Amersham). The maximum gray value uptake in the tissue block was converted into % ID per area based on the diluted tracer standards using ImageJ 1.53k. The bar in the H&E and PSMA IHC images represents 1000 mm.

H&E, hematoxylin and eosin; IHC; immunohistochemistry; PSMA, prostate-specific membrane antigen

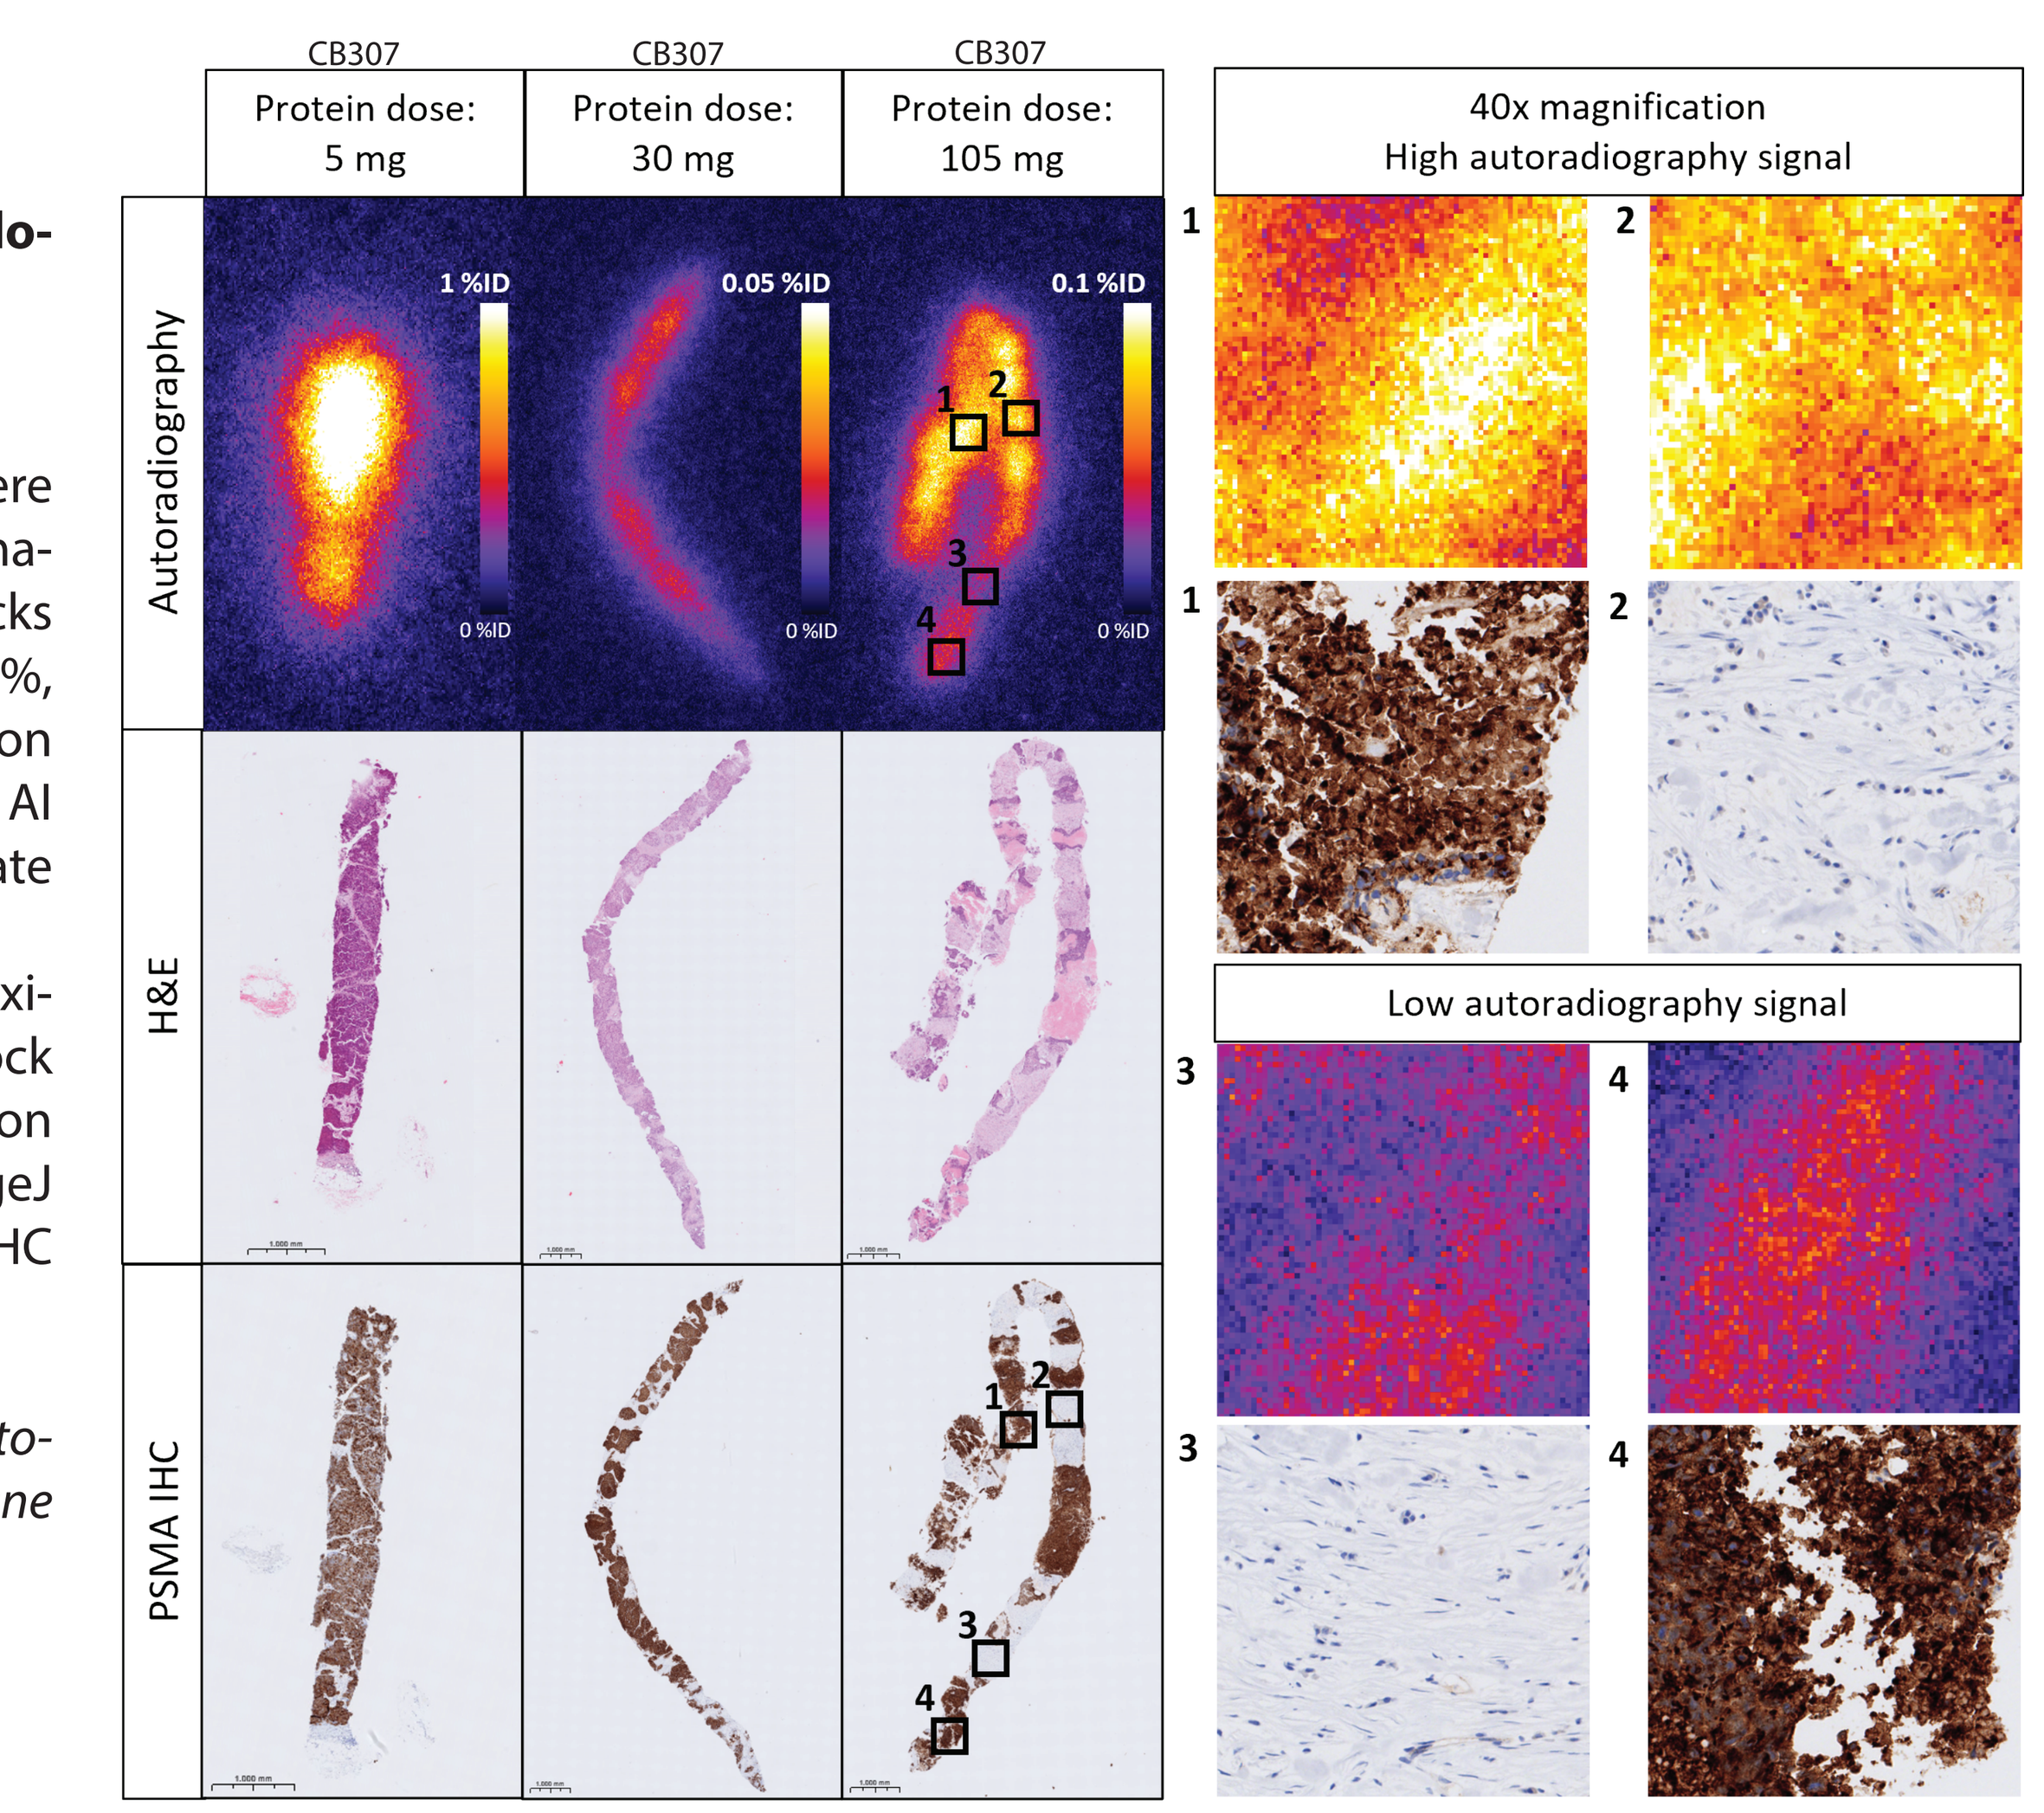
CB307
Protein dose: 5 mg
CB307
Protein dose: 30 mg
CB307
Protein dose: 105 mg
40x magnification
High autoradiography signal
Autoradiography
1 %ID
0 %ID
0.05 %ID
0 %ID
0.1 %ID
0 %ID
H&E
PSMA IHC
Low autoradiography signal
1
2
3
4

Conclusion

Trispecific Humabody® V_H CB307 targets PSMA only in tumors, without specific uptake in PSMA-positive normal tissues or lymphoid tissues.

D.G. Knapen reports Institutional Financial Support for clinical trials / contracted research from Bayer, outside the submitted work.

This study is sponsored by Crescendo Biologics.

Contact this researcher via: d.g.knapen@umcg.nl